Antimicrobial activity of 2-tert-butylhydroquinone (TBHQ) and bismuth or copper, new synergistic combinations for the treatment of acne

Daniel J Fitzgerald, Priscila Gottardello, Scott Seville, E Anne Eady, Jon H Cove

Syntopix Group plc, Institute of Pharmaceutical Innovation, Bradford, BD7 1DP, UK

P101

TOPICAL ANTIMICROBIAL INNOVATION

Abstract

Acne affects a majority of the adolescent and young adult population. It is a multifactorial disorder associated with *Propionibacterium acnes* infection of functionally blocked pilosebaceous follicles. Commonly treated with topical antibiotics, concerns regarding increasing resistance have led to the need for alternative anti-infectives. 2-*t*-butylhydroquinone (TBHQ) is a lipophilic sterically hindered hydroquinone commonly used as an antioxidant in food. The antimicrobial properties of TBHQ against food-related bacteria are well known but the compound's pronounced activity against propionibacteria is a recent discovery. Under appropriate conditions, TBHQ can be oxidised to 2-*t*-butylbenzoquinone (TBBQ) with which it exists in a redox couple. The oxidation reaction proceeds via a semiquinone intermediate resulting in free radical generation. At neutral to acidic pHs, the reaction occurs slowly but can be catalysed by transition metal ions such as copper. In order to determine whether this mechanism could boost the antimicrobial activity of TBHQ, we compared the potency of TBHQ in the presence and absence of selected transition and other metal salts. Using a modified disk diffusion assay (DDA), TBHQ in combination with either copper sulphate (CuSO4), or bismuth subsalicylate (BiSS), a non-transition metal salt, markedly increased the mean zone of inhibition (ZoI) above that of the most active compound alone against *Propionibacterium acnes* NCTC737. Mean ZoIs from triplicate assays increased by 11.68 mm (50.3% area increase) in combination with CuSO4 and by 10.83 mm (129.6% area increase) with BiSS. Similar levels of potentiation in modified DDAs were observed using different copper and bismuth salts and in a non-predictable way with certain other metal salts. The potentiation of TBHQ with either CuSO4 or BiSS versus *P. acnes* was confirmed against a panel of antibiotic susceptible and resistant propionibacterial strains. Checker-board fractional inhibitory concentration (FIC) assays showed the interaction of TBHQ with CuSO4 or BiSS against *P. acnes* to be synergistic with lowest FIC indices of 0.5 and 0.31 respectively (synergy = FICI ≤0.5). We conclude that synergy versus *P. acnes* is not confined to transition metal interactions with TBHQ nor is it a general property of transition metals that catalyse redox cycling. Synergistic combinations of TBHQ with copper or bismuth offer a novel acne treatment against which resistance would be very unlikely to develop.

Background

- TBHQ is a sterically hindered phenolic which is a potent lipophilic antioxidant used in food (E319) and cosmetics[1]
- The antimicrobial properties of TBHQ against food-related bacteria are well known[2]; pronounced activity against propionibacteria has only recently been demonstrated
- Antimicrobial activity of TBHQ is in part due to disruption of membrane function/integrity[3]
- Under certain conditions, TBHQ exhibits pro-oxidant effects.
- Metal ions catalyse the oxidation of TBHQ to TBBQ with concomitant generation of ROS & regeneration of TBHQ via redox cycling.[4,5]
- ROS are inhibitory to bacteria.
- Increased production of ROS may enhance the ability of TBHQ to inhibit bacteria.

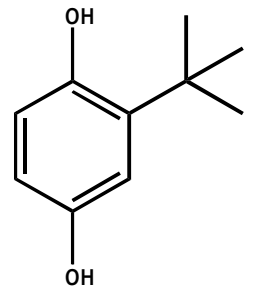
OH
OH

2-tert-butylhydroquinone (TBHQ)

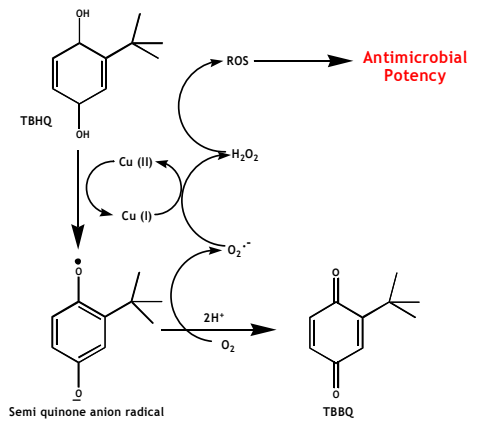
TBHQ
ROS
Antimicrobial Potency
H_2O_2
Cu (II)
Cu (I)
$O_2^{\cdot-}$
2H+
O_2
Semi quinone anion radical
TBBQ

Objective

- To determine whether the combination of a Cu or a Bi salt with TBHQ can potentiate antimicrobial activity versus *Propionibacterium acnes.*

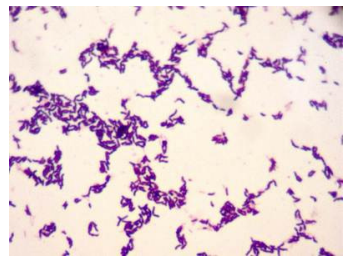

P. acnes

Methods

- A modified disk diffusion assay was used to identify potential synergistic combinations of TBHQ with either Cu or Bi salts vs *Propionibacterium acnes* NCTC 737.
- The potentiation of TBHQ with either Cu or Bi salts was confirmed against a panel of 24 antibiotic susceptible and resistant propionibacterial strains.
- Checker-board fractional inhibitory concentration (FIC) assays were used to quantify synergistic interactions.

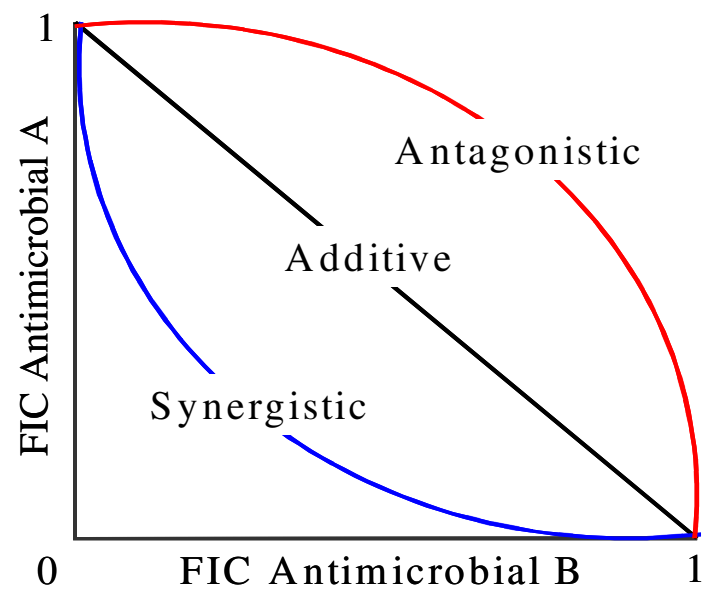
1
0
1
FIC Antimicrobial A
FIC Antimicrobial B
Antagonistic
Additive
Synergistic

MIC data transformed into a fractional inhibitory concentration.
FICA = MIC of A with B / MIC of A

FIC index = FICA + FICB :
0.51 - 4 = Indifferent
≤0.5 = Synergy
≥4.0 = Antagonism

Activity of copper & bismuth salts in combination with TBHQ verus *P. acnes* NCTC 737 in disk diffusion assays

TBHQ + metal salt	ZoI increase (mm)	Area increase (%)
Copper (II) sulphate	11.68	50.3
Copper (II) 8-hydroxyquinoline	7.93	111.6
Copper (I) bromide	15.44	94.3
Copper (I) iodide	21.48	107.9
Copper (II) acetate	13.35	56.0
Copper (II) bromide	11.47	46.2
Copper (II) nitrate	15.85	69.6
Copper (I) acetate	18.67	84.6
Copper (II) oxychloride	8.24	34.8
Copper (II) sulphate pentahydrate	22.11	108.6
Copper (II) D-gluconate	8.13	39.1
Copper (II) salicylate	27.84	150.5
Copper (II) aspirinate	16.48	88.2
Bismuth subsalicylate	10.83	129.6
Bismuth acetate	6.72	53.4
Bismuth chloride	5.57	47.3
Bismuth citrate	8.94	58.6

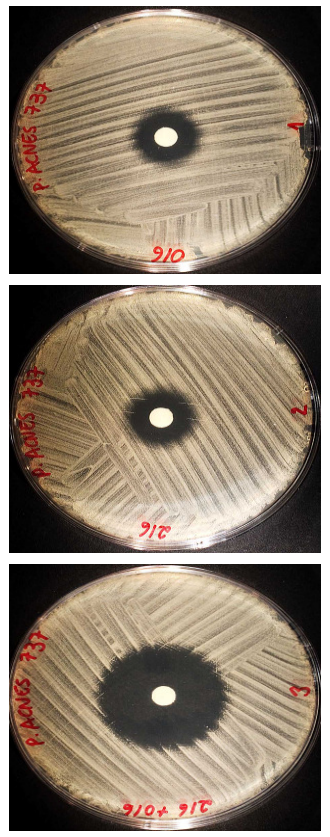

A

B

A + B

Activity of CuSO4 & bismuth subsalicylate in combination with TBHQ versus antibiotic susceptible & resistant* cutaneous propionibacteria in disk diffusion assays

Combination	Species	Nº of strains	ZoI increase (mm) – range (mean)	Area increase (%) – range (mean)
TBHQ + $CuSO_4$	*P. acnes*	16	7.63 - 30.53 (18.2)	28.09 - 189.65 (92.9)
	P. granulosum	8	5.85 - 14.53 (11.5)	21.77 - 156.55 (99.0)
TBHQ + bismuth subsalicylate	*P. acnes*	16	6.21 - 14.29 (9.3)	70.15 - 294.23 (135.4)
	P. granulosum	8	4.56 - 9.94 (7.8)	55.61 - 153.56 (114.7)

* *P. acnes* with no resistance (1), Tet/MLS (5), Tet (2), Clin (2), Ery (1), MLS (3) and MLSK (2); *P. granulosum* with no resistance (2), MLS (4) and MLSK (2).

CuSO4 & bismuth subsalicylate interact synergistically with TBHQ

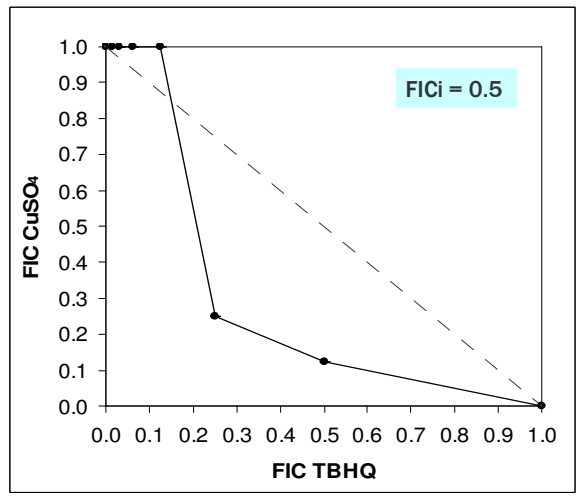
FICi = 0.5
FIC $CuSO_4$
FIC TBHQ
0.0
0.1
0.2
0.3
0.4
0.5
0.6
0.7
0.8
0.9
1.0

FIC isobologram for TBHQ + Cu SO_4 against *P. acnes* NCTC 737.

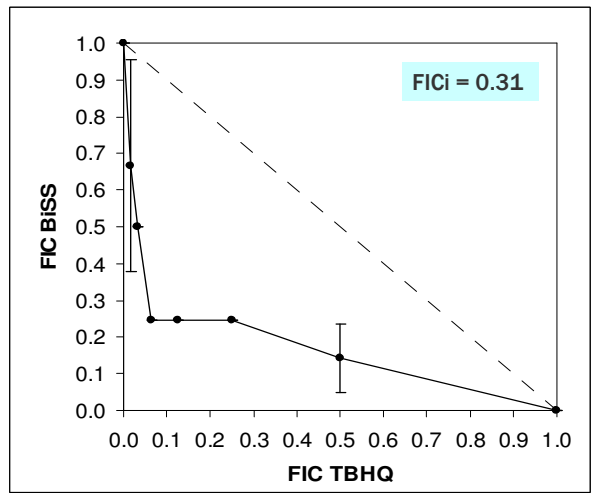
FICi = 0.31
FIC BiSS
FIC TBHQ
0.0
0.1
0.2
0.3
0.4
0.5
0.6
0.7
0.8
0.9
1.0

FIC isobologram for TBHQ + Bismuth subsalicylate against *P. acnes* NCTC 737.

Summary & Conclusions

- TBHQ has a history of safe use in man, including low irritant potential when applied topically (data on file).
- Synergistic combinations of TBHQ with either a copper or a bismuth salt offer a novel and potentially well tolerated acne treatment against which resistance would be unlikely to develop.

References

1. Cosmetic Ingredient Review. Final report on the safety assessment of tert-butylhydroquinone. *J Am Coll Toxicol* 1986; 5: 329-51.
2. Fung DY et al. Effect of phenolic antioxidants on microbial growth. *Crit Rev Microbiol* 1985; 12: 153-83.
3. Keweloh H, Weyrauch G, Rehm HJ. Phenol-induced membrane changes in free and immobilised *Escherichia coli*. *Appl Microbiol Biotechnol* 1990; 33: 66-71.
4. Li Y et al. Copper redox-dependent activation of 2-tert-butyl(1,4)hydroquinone: formation of reactive oxygen species and induction of oxidative DNA damage in isolated DNA and cultured rat hepatocytes. *Mutat Res* 2002; 518: 123-33.
5. Okubo T et al. Contribution of oxygen radicals to DNA cleavage by quinone compounds derived from phenolic antioxidants, tert-butylhydroquinone and 2,5-ditert butylhydroquinone. *Toxicol Lett* 1997; 90: 11-18.

American Association of Dermatology Summer Meeting 2009

www.syntopix.com